PROACT ▶ A new way of engaging and empowering patients that fundamentally changes our understanding of how tolerability impacts in early clinical development.

Andrew Hughes[1], Donal Landers[2], Tobias Arkenau[3], Jennifer Royle[2]

[1] Manchester Cancer Research Centre, University of Manchester, Manchester, UK; [2] AstraZeneca, Alderley Park, Macclesfield, UK; [3] Sarah Cannon Research Institute, London, UK.

P160

Background

- We have developed PROACT (Patient Reported Opinions About Clinical Tolerability), a mobile application designed to empower patients participating in early clinical studies.
- It is a unique, patient-centric approach to engaging and working with patients as partners in a clinical study.
- PROACT allows the patient to create a portable 'video diary' and two-way video communication between the patient and their clinical team. It also gives patients the unique opportunity and choice to share with the sponsor their insights and thoughts on an investigational agent and contribute to drug development.

Objective

- To investigate patient uptake of PROACT and to characterise information captured by the PROACT system.

Methods

Study design, participants and data collection

- Patients recruited to Phase I AstraZeneca (the Sponsor) oncology studies at the Sarah Cannon Research Institute, London, UK were eligible to participate if they had a "smart" phone or tablet computer and were willing to provide informed consent.
- Messages (audio and/or video) were streamed to a server and made instantly viewable to the onsite clinical team who responded appropriately.
- After 32 days (which allowed the patients and their onsite clinical team to discuss the content of the audio/video), personally identifiable information was removed from the messages by a small group of analysts before being sent to the Sponsor's Study Team.

Data analysis

- A hierarchical dictionary and flexible coding database were created to allow for aggregation of data, whilst linking back to the patient's own words, and for every topic mentioned, the outcome and impact were captured (if reported).
- A simple colour-scale was developed to visualise the impact of the reported topic upon that patient's wellbeing or daily activities.

Ethics approval

- The study received Ethical Research Committee approval and was conducted in accordance with applicable regulatory requirements and other relevant guidelines, such as patient data privacy requirements.

Results

- Of the 16 patients informed of the PROACT initiative, 8 patients used a "smart" phone or computer tablet and consented to take part.
 - Participating patients were marginally younger (mean age 61 yrs; range 45–68 yrs) than those ineligible to participate due to lack of access to mobile technology (mean age 67 yrs; range 51–80 yrs).
- Use of PROACT varied and all messages volunteered by patients were relevant and informative for drug development.
 - Topics covered included impact of safety and tolerability, relay of self-monitored results (e.g. blood pressure), feedback on study design, and formulations (see Table 1).

Table 1. Examples of patient feedback received by topic

Topic	Example feedback
Safety	Patient was monitoring own blood pressure and sending results back to clinical team in between visits. Example quotes include: *"I spent Sunday in bed shaking off a headache and feeling unwell."* *"I'm not doing an awful lot because of the fatigue that is really quite, quite overwhelming."*
Convenience	*"The taste of the pills is unpleasant even though they are in the mouth for only a couple of seconds, ... if it was possible to mask the taste of the pills that might help."*
Study design	*"I have been fairly tired as this is second day after chemo and the fairly demanding day of giving blood for the trial."*

- Patient feedback via PROACT provided an extra level of insight. Alignment with the clinical study data provided a richer understanding of tolerability, the impact on patients and the overall consequences (see Table 2).

Table 2. Example comparison of clinical and PROACT data relating to the topic

Data source	Information recorded/feedback provided
Clinical study database	MedDRA[1] term 'Rash', CTCAE Grade 1, ongoing.
PROACT	*"The rash is gone from my face and is greatly reduced on my chest."* *"It is also quite unpleasant to expose my skin to hot water – I don't enjoy a shower or a bath."* *"My skin continues to be dry but is responding to stuff called Bach's rescue cream and is not a problem."*

[1] MedDRA: Medical Dictionary for Regulatory Activities.

Figure 1. Analysis and graphical representation of patient feedback.

Quote type summary

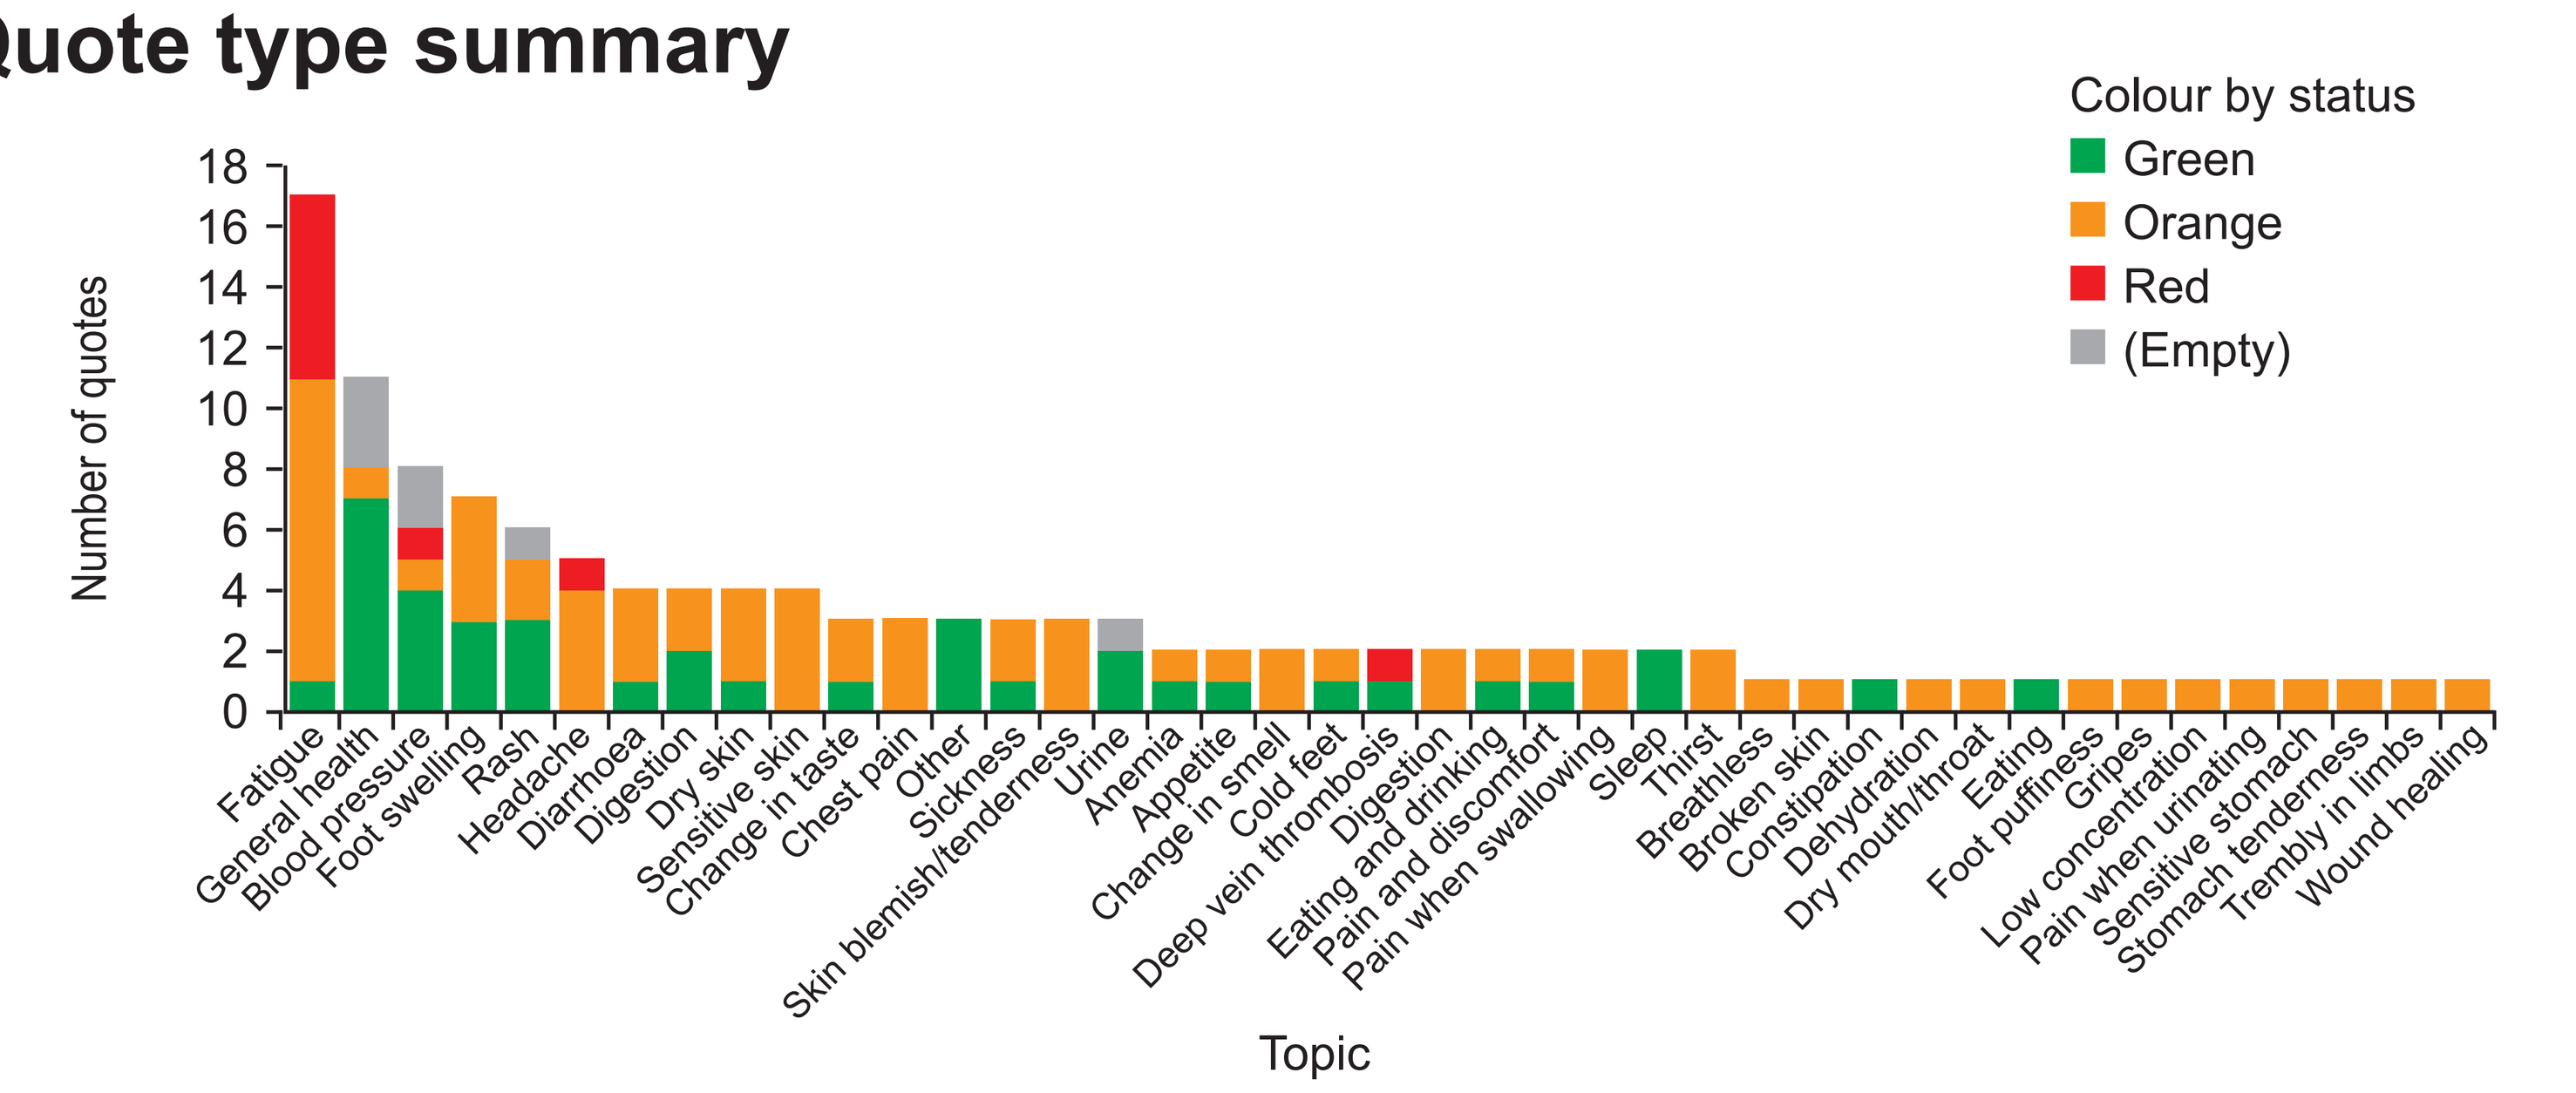
Colour by status
Green
Orange
Red
(Empty)
Number of quotes
0 2 4 6 8 10 12 14 16 18
Fatigue
General health
Blood pressure
Foot swelling
Rash
Headache
Diarrhoea
Digestion
Dry skin
Sensitive skin
Change in taste
Chest pain
Other
Sickness
Skin blemish/tenderness
Urine
Anemia
Appetite
Change in smell
Cold feet
Deep vein thrombosis
Digestion
Eating and drinking
Pain and discomfort
Pain when swallowing
Sleep
Thirst
Breathless
Broken skin
Constipation
Dehydration
Dry mouth/throat
Eating
Foot puffiness
Gripes
Low concentration
Pain when urinating
Sensitive stomach
Stomach tenderness
Trembly in limbs
Wound healing
Topic

Reported impact

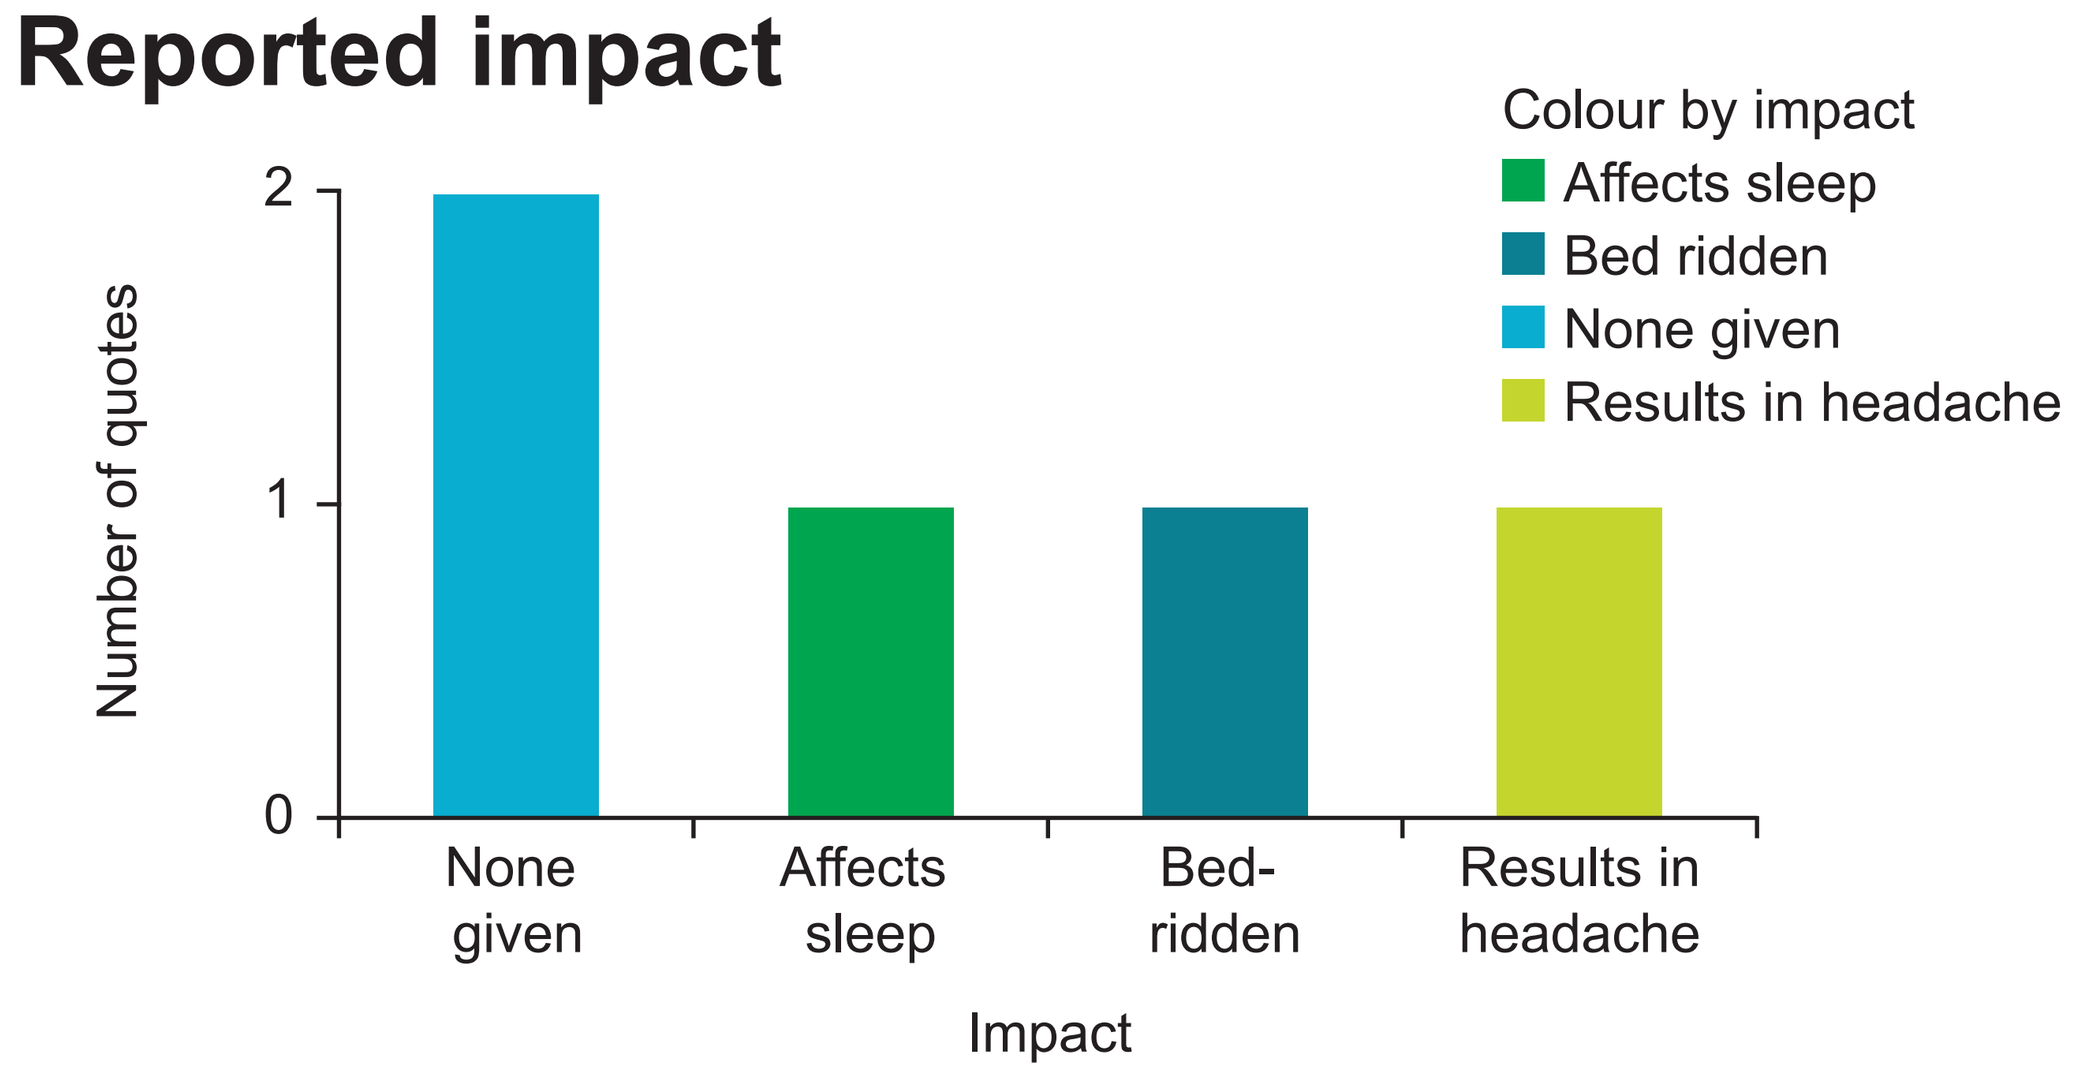
Colour by impact
Affects sleep
Bed ridden
None given
Results in headache
Number of quotes
0 1 2
None given
Affects sleep
Bed-ridden
Results in headache
Impact

Response/Outcome

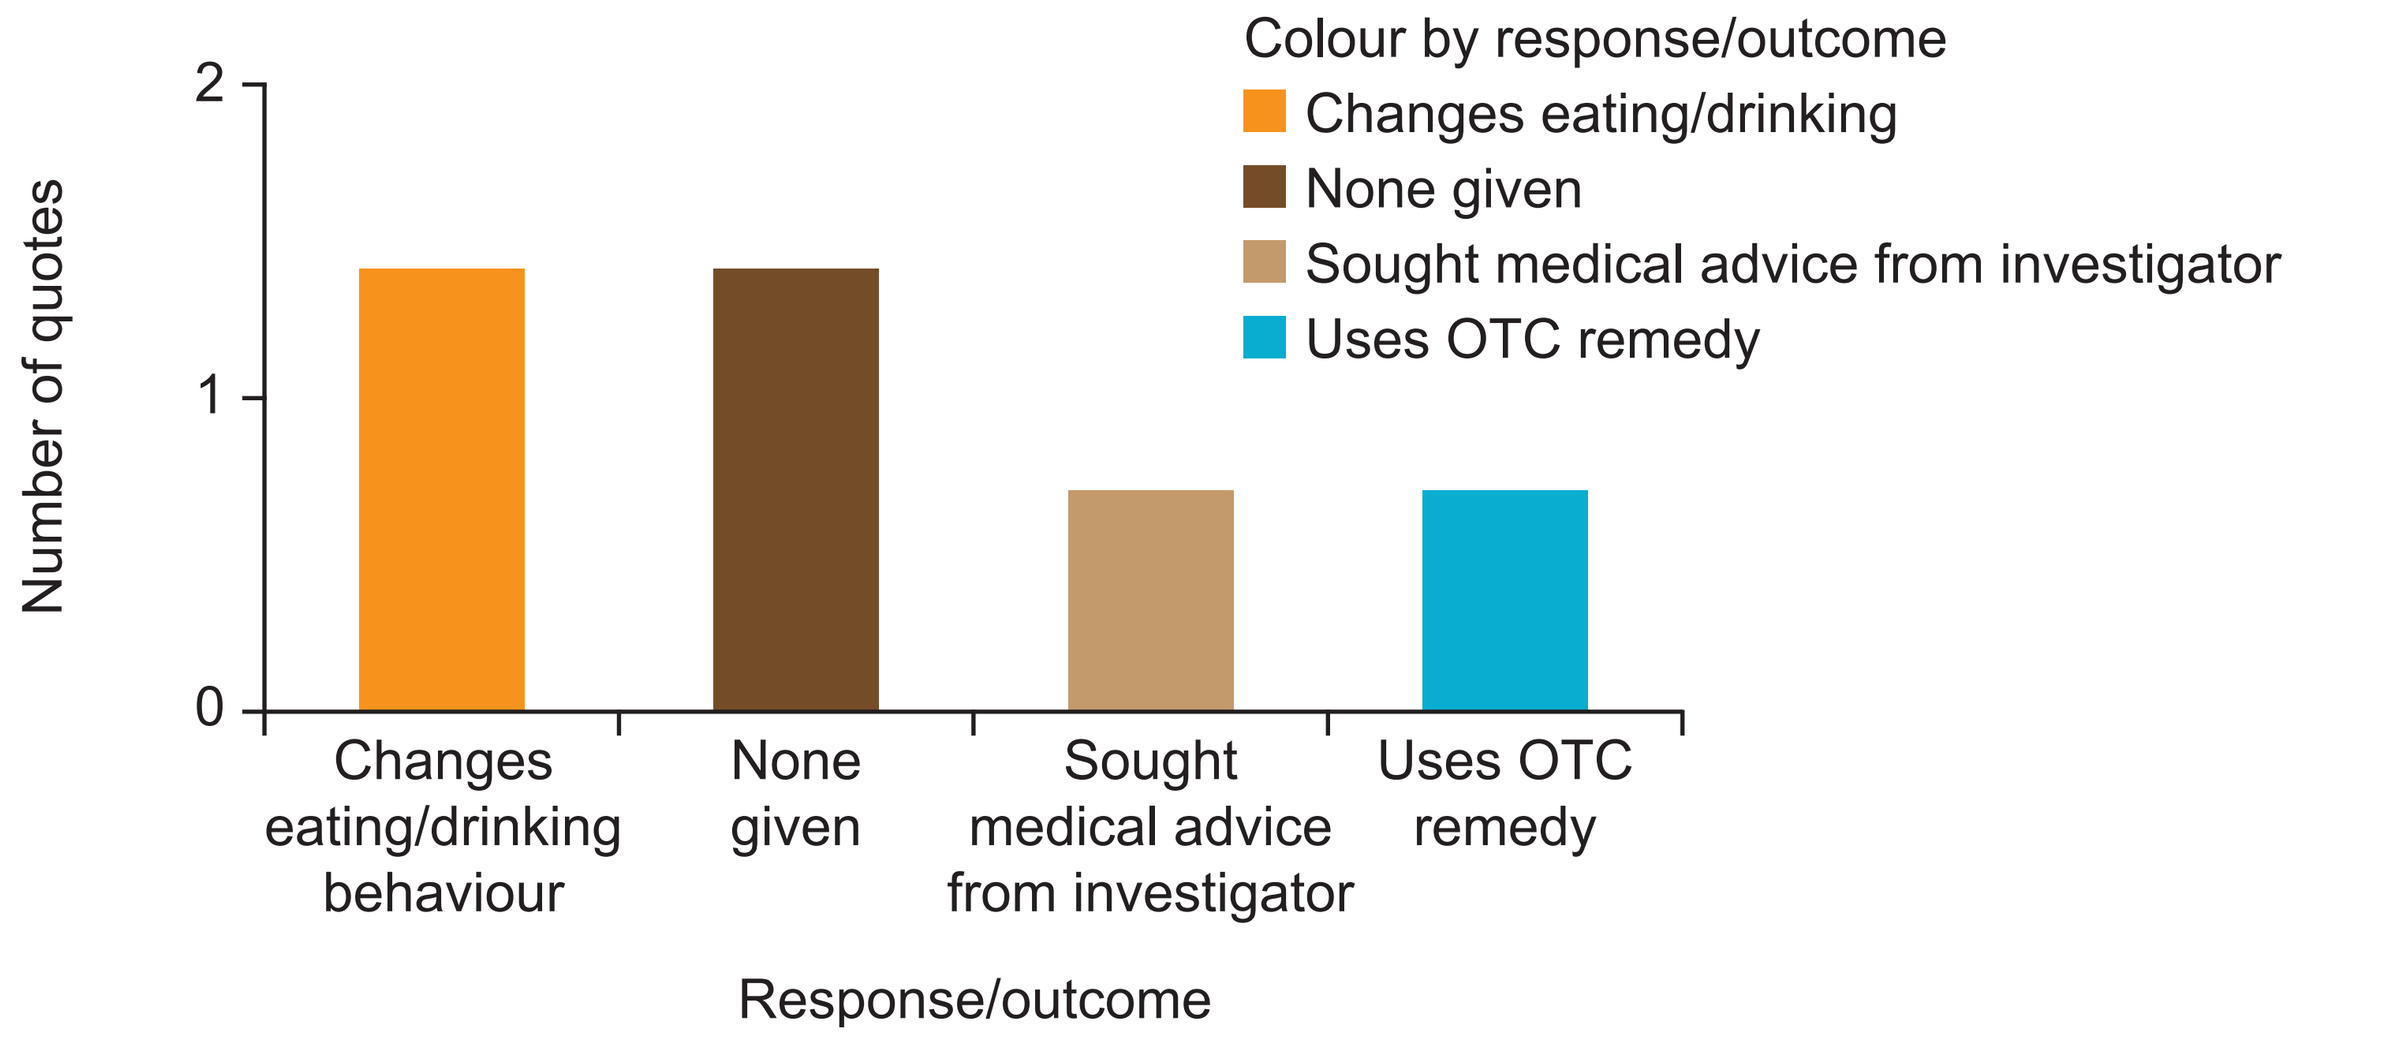
Colour by response/outcome
Changes eating/drinking
None given
Sought medical advice from investigator
Uses OTC remedy
Number of quotes
0 1 2
Changes eating/drinking behaviour
None given
Sought medical advice from investigator
Uses OTC remedy
Response/outcome

Quotes over time

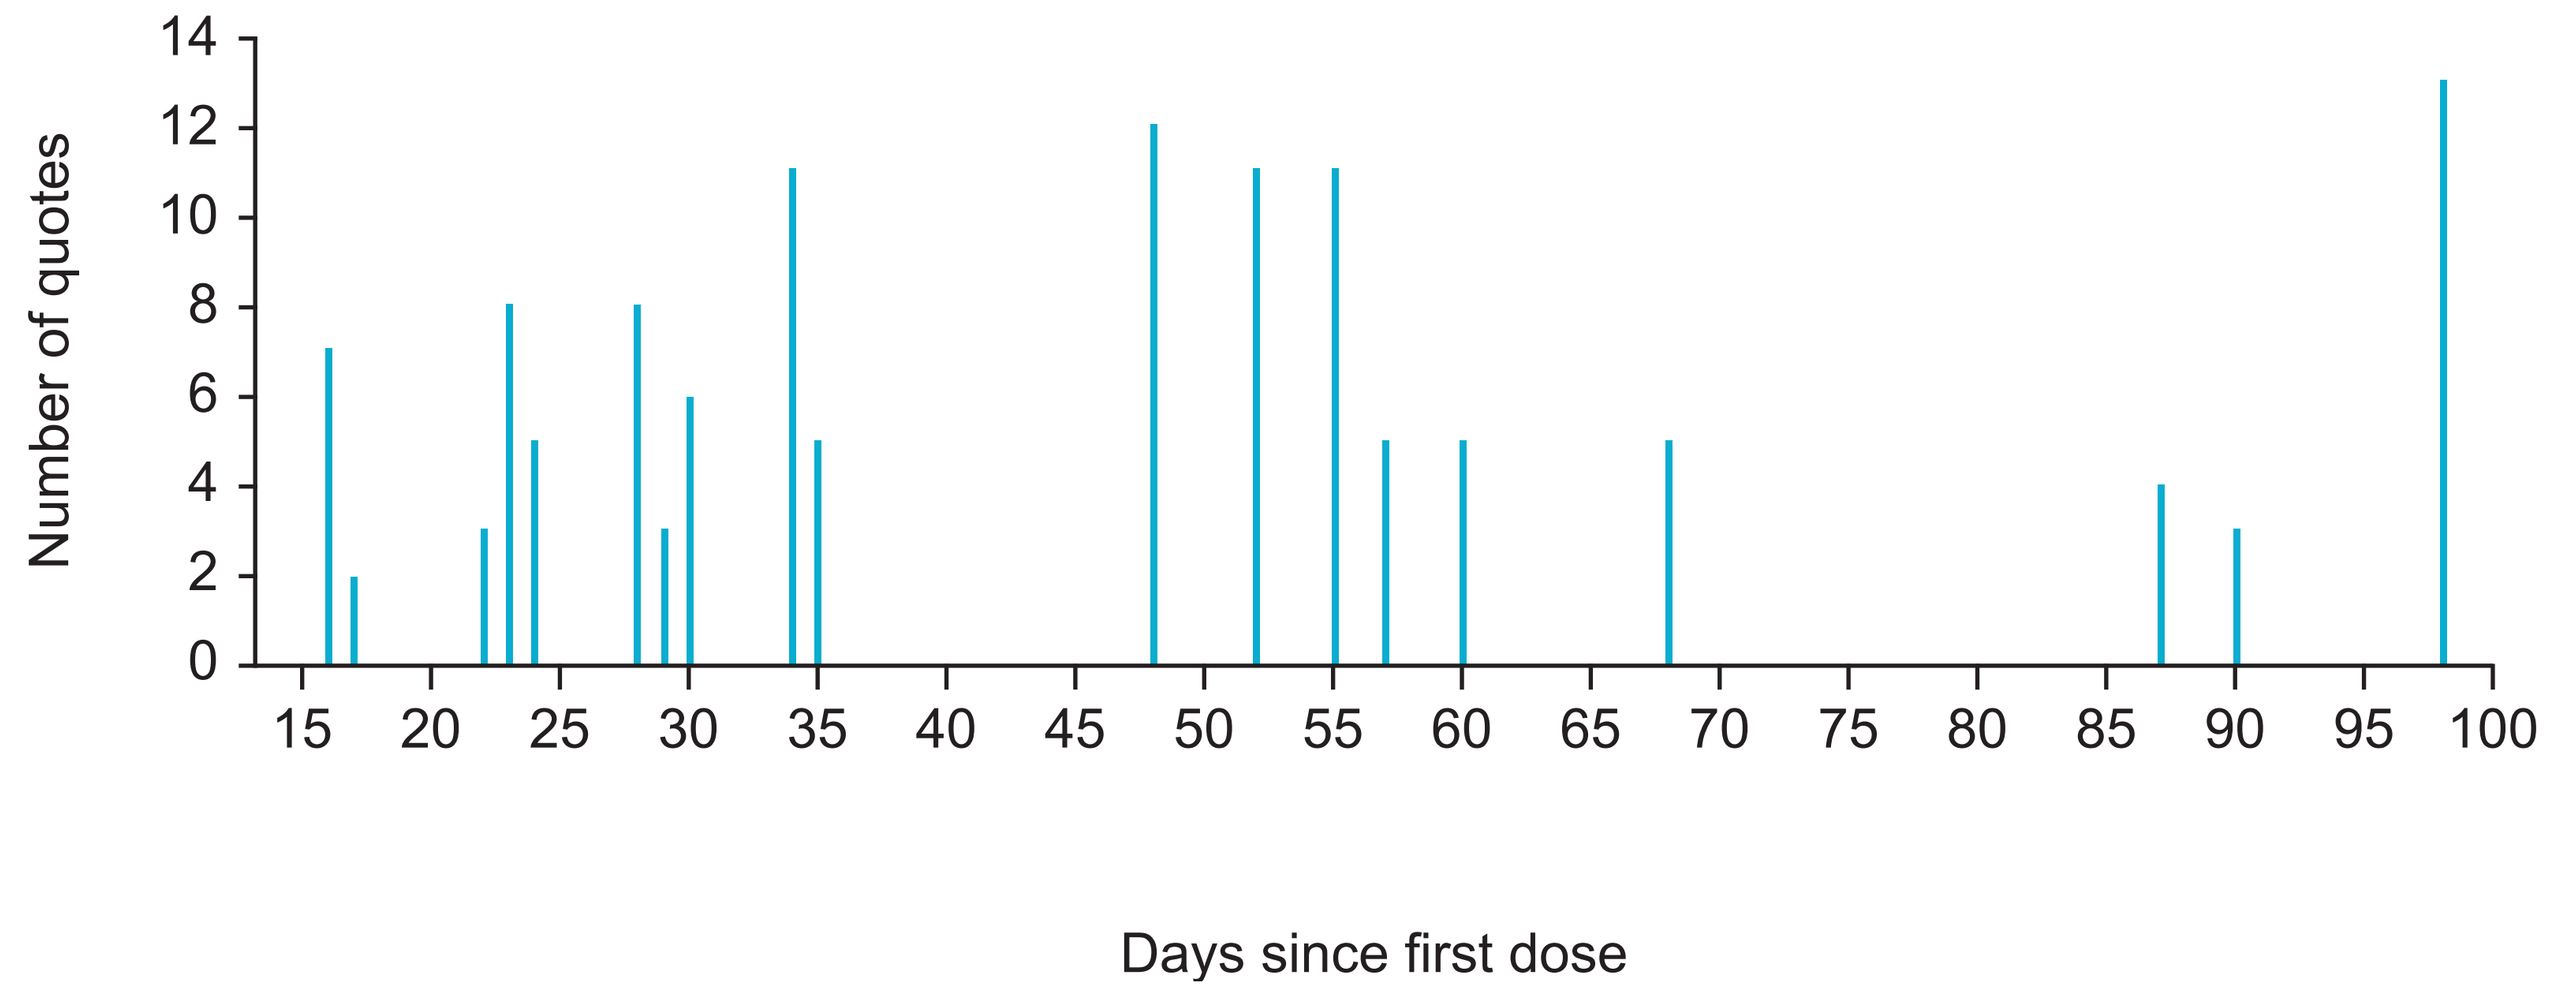
Number of quotes
0 2 4 6 8 10 12 14
15 20 25 30 35 40 45 50 55 60 65 70 75 80 85 90 95 100
Days since first dose

- The coded database was visualised in a series of graphs to explore potential signals (Figure 1).
- The results of the analysis enabled the Sponsor to begin to answer questions such as '*which aspects of study treatment or conduct are most impactful*?', and '*how are patients self-managing their care*?'.
 - Not only does this inform drug-development strategy, it also creates the potential for sharing this understanding back with the clinical teams across the whole study to provide patient support and to enhance patient experience.

Acknowledgments

This study was sponsored by AstraZeneca. Medical writing support, funded by AstraZeneca, was provided by David Peters MSc of Sequoia Medical Communications. The authors acknowledge the contribution of all the patients who took part in the study, colleagues within the AstraZeneca development team (most notably in Amrik Mahal and Åsa Strom from Research and Development Information, Stephanie Roberts from the Advanced Analytics Centre, and Saj Shah from the Legal Department), Gabriel Mak of the Sarah Cannon Research Institute for help with identifying patients, and Bartley O'Connor of PatientPharma Ltd for development of the application used in this pilot project.

Conflict of interest

Donal Landers, and Jennifer Royle are employees of AstraZeneca. Andrew Hughes was an AstraZeneca employee at time of study completion.

Conclusions

- This completed pilot study showed that PROACT is a valid approach to engage and empower patients in early oncology clinical trials and to gain further insight into their experience beyond that documented in the traditional clinical data set.
- This novel approach has the potential to fundamentally change the communication paradigm with patients within an ongoing clinical trial.
 - As previous studies have shown, engaged patients who have the skills, ability and willingness to help manage their own health have better health outcomes at lower cost to the health system.[1]

Reference

1. Creating new value with patients, carers aand communities. KPMG International 2015 http://www.kpmg.com/Global/en/IssuesAndInsights/ArticlesPublications (accessed 19th August 2015)

Supported by AstraZeneca IMED Biotech Unit

18th ECCO – 40th ESMO European Cancer Congress, Messe Wien Exhibition and Congress Centre, 1021 Vienna, Austria, September 25–29, 2015